Second-line afatinib for patients with locally advanced or metastatic NSCLC harbouring common *EGFR* mutations: a Phase IV study

#157P

Sumitra Thongprasert,[1]* Sarayut L. Geater,[2] Dana Clement,[3] Amr Abdelaziz,[4] Jasmin Reyer-Igama,[5] Dragana Jovanovic,[6] Sheethal Suresh,[7] Agnieszka Cseh,[8] Rabab Gaafar[9]

[1]Medical Oncology Unit, Wattanosoth Hospital and Bangkok Hospital Chiang Mai (BDMS), Chiang Mai, Thailand; [2]Department of Internal Medicine, Songklanagarind Hospital, Prince of Songkla University, Songkhla, Thailand; [3]Medical Oncology Department, Regional Oncology Institute of Iasi, Iasi, Romania; [4]Clinical Oncology Department, Alexandria Clinical Research Centre, Alexandria University, Alexandria, Egypt; [5]Department of Internal Medicine, Hematology and Oncology, Baguio General Hospital and Medical Center, Baguio City, Philippines; [6]Clinic for Pulmonology, Clinical Center Serbia, Belgrade, Serbia; [7]Oncology Department, Boehringer Ingelheim Pharmaceuticals, Inc., Singapore, Republic of Singapore; [8]Medical Department, Boehringer Ingelheim RCV GmbH & Co. KG, Vienna, Austria; [9]Medical Oncology Department, National Cancer Institute, Cairo University, Cairo, Egypt

Introduction

- The oral, irreversible ErbB family blocker afatinib is approved in many countries for the treatment of *EGFR* mutation-positive (*EGFR*m+) locally advanced or metastatic NSCLC
 - The recommended starting dose of afatinib is 40 mg/day
- Afatinib improved PFS versus chemotherapy in the LUX-Lung 3 and 6 trials,[1,2] and versus gefitinib in LUX-Lung 7[3] in patients with *EGFR*m+ NSCLC receiving first-line afatinib 40 mg/day
- The LUX-Lung 2 study[4] supports the use of second-line afatinib in patients with *EGFR*m+ NSCLC progressing following first-line platinum-based chemotherapy, but only 7/68 (10%) patients received afatinib 40 mg/day; most patients (90%) received afatinib 50 mg/day
 - As such, data supporting the use of afatinib 40 mg/day as second-line therapy in patients with *EGFR*m+ NSCLC are limited

PFS, progression-free survival

Objectives

- The aim of this trial was to assess the efficacy and safety of afatinib 40 mg/day as second-line therapy in patients with advanced/metastatic NSCLC harboring *EGFR* common mutations (Del19 and/or L858R), who had progressed following first-line platinum-based chemotherapy and were EGFR TKI-naïve

Methods

- Multicenter, open-label, single-arm Phase IV study conducted across 24 sites in 7 countries (Egypt, Malaysia, Philippines, Poland, Romania, Serbia, and Thailand)

Key inclusion criteria

- Aged ≥18 years
- Del19 and/or L858R
- ECOG PS 0 or 1
- Confirmed stage IIIB or IV lung adenocarcinoma
- Radiologically confirmed progression or recurrence following first-line chemotherapy

Key exclusion criteria

Previous treatment with:

- More than 1 prior line of therapy*
- Less than 3 cycles of platinum-based chemotherapy
- Any EGFR-targeting TKI or antibody
- Any treatment within 3 weeks prior to the start of trial
- Major surgery within 4 weeks before starting trial

*Not counting radiotherapy, radiosensitisers, and/or intrapleural administration of anti-cancer agents.
ECOG PS, Eastern Cooperative Oncology Group performance status

Methods (Cont'd)

- Eligible patients received afatinib 40 mg/day
- If patients developed grade ≥3 drug-related adverse events (AEs), grade 2 diarrhoea for more than 2 days, or rash for more than 7 days, treatment was interrupted until recovery to grade ≤1, followed by dose reduction in 10 mg decrements
- Patients were treated with afatinib until disease progression or discontinuation for other reasons

Endpoints

Endpoint	
Primary	• OR* (RECIST v1.1)
Secondary	• Disease control† • PFS
Safety	• Intensity and incidence of AEs‡

*Defined as CR or PR; †Defined as CR, PR, or SD; ‡Graded according to US National Cancer Institute Common Terminology Criteria for Adverse Events (NCI CTCAE) Version 3.0

AE, adverse event; CR, complete response; OR, objective response; PFS, progression-free survival; PR, partial response; RECIST, Response Evaluation Criteria In Solid Tumors; SD, stable disease

- Efficacy and safety were evaluated in a descriptive manner, and there were no formal statistical hypotheses

Results

Patients and treatment

- 60 patients received at least one dose of afatinib 40 mg/day (Table 1)
- 86.7% of patients were treated for at least 90 days; mean treatment duration was 336.7 days

Table 1. Patient disposition.

	Afatinib 40 mg/day	
	n	%
Enrolled	70	
Treated	60	
Patients discontinued from afatinib	60	100.0
Progressive disease according to RECIST	24	40.0
Clinical signs and symptoms of progression	2	3.3
AEs	12	20.0
Non-compliant with protocol	1	1.7
Lost to follow-up	0	0.0
Refused to continue afatinib	1	1.7
Other*	20	33.3

*Switched to commercially-available afatinib

Results (Cont'd)

Patient demographics and baseline characteristics

- Patients were mostly white (68.3%) and had a mean age (Std dev) of 59.9 (9.8) years
- The most frequent sites of distant metastases were the ipsilateral lung (48.3%) and contralateral lung (45.0%), while 15.0% of patients had brain metastases at screening
- All patients had prior chemotherapy, while 17 (28.3%) and 18 (30.0%) had surgery and radiotherapy, respectively

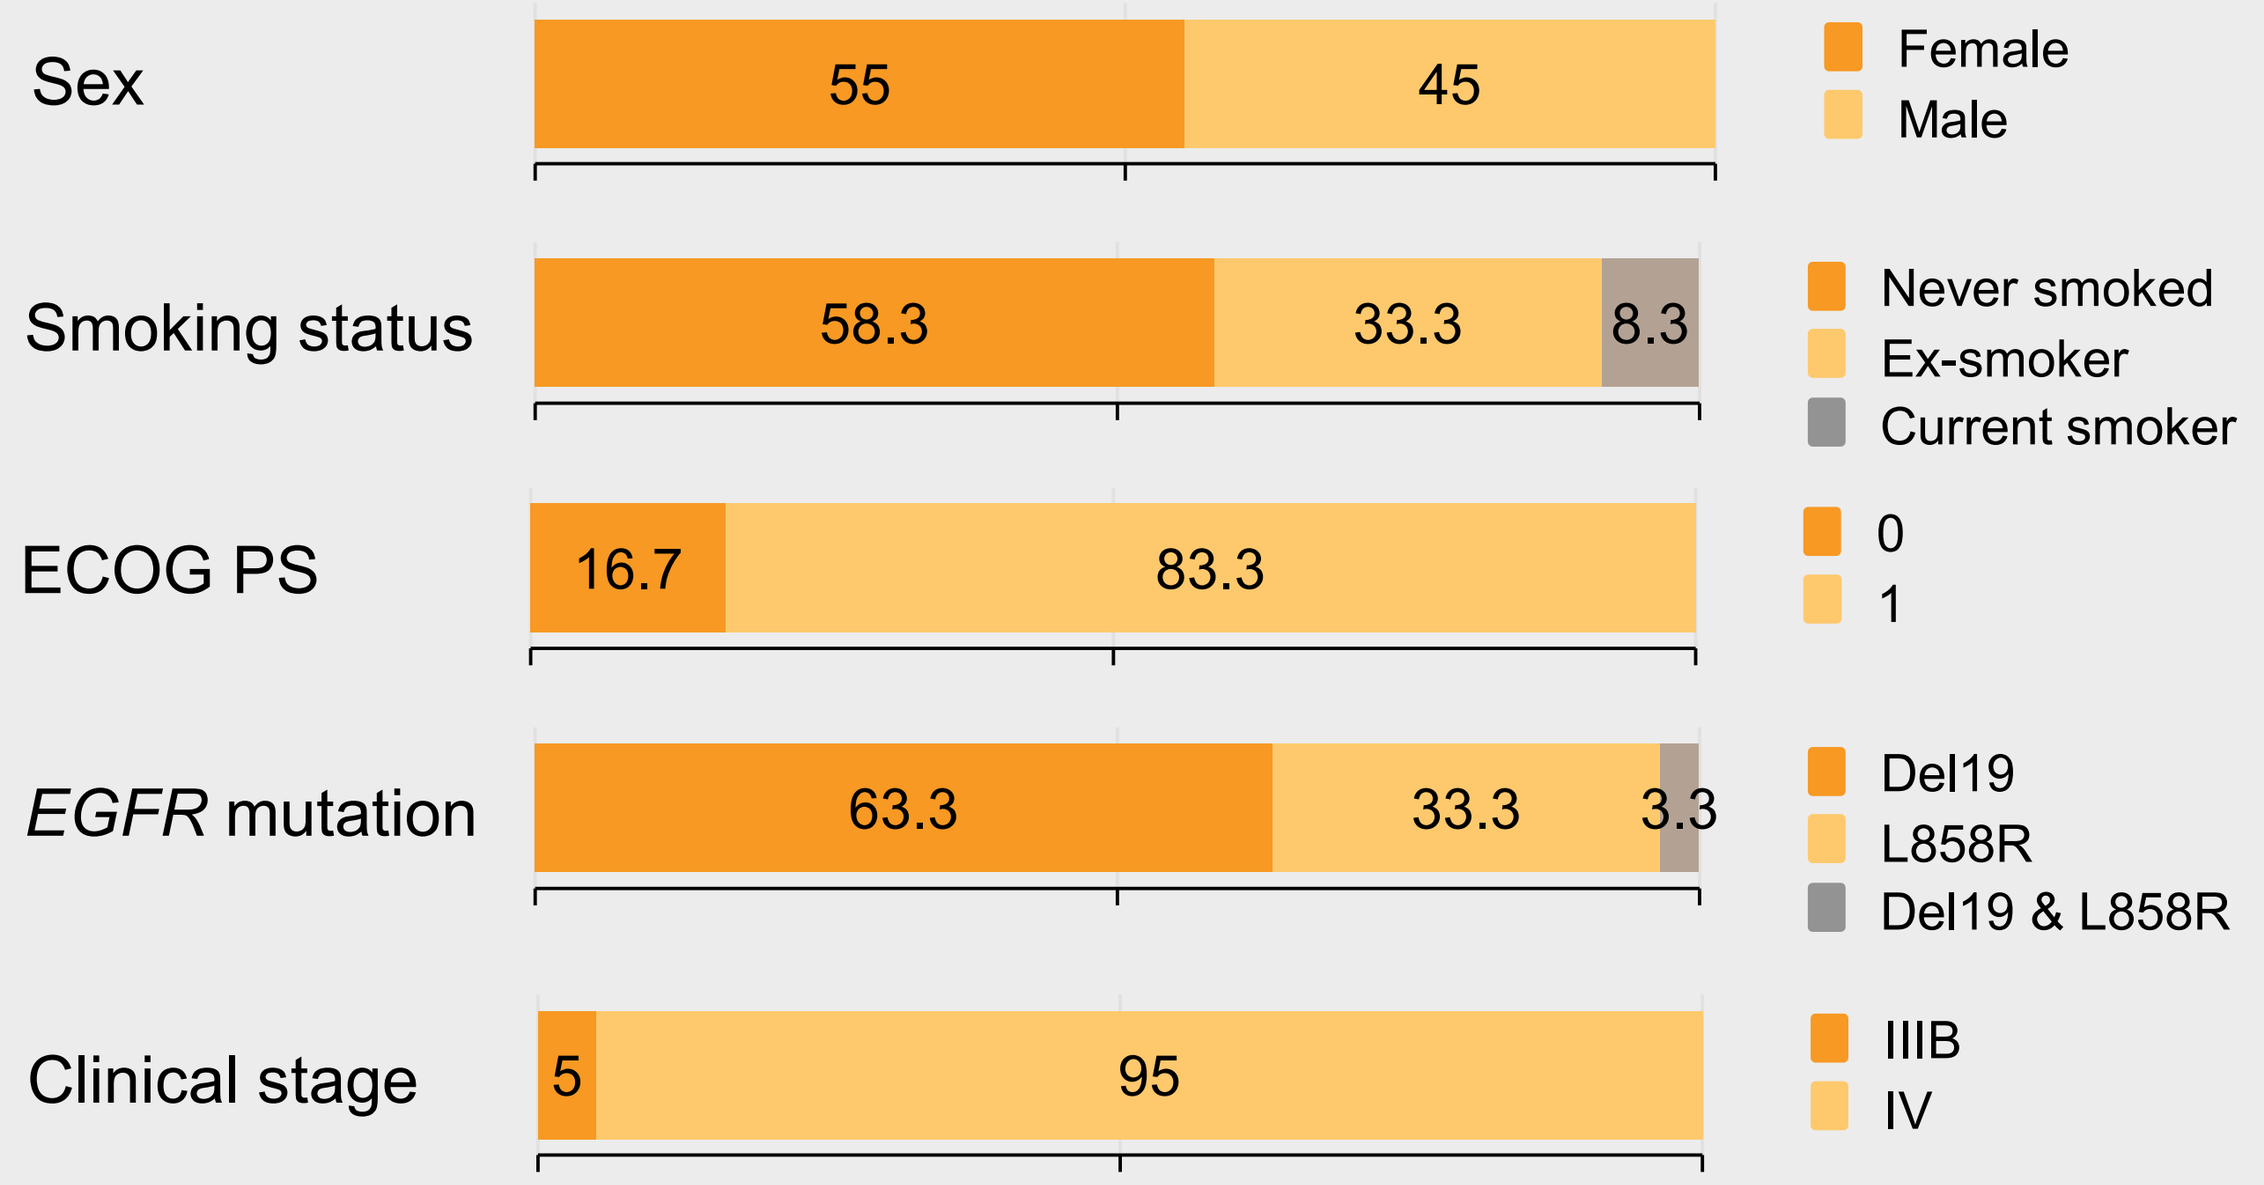
Sex
55
45
Female
Male
Smoking status
58.3
33.3
8.3
Never smoked
Ex-smoker
Current smoker
ECOG PS
16.7
83.3
0
1
EGFR mutation
63.3
33.3
3.3
Del19
L858R
Del19 & L858R
Clinical stage
5
95
IIIB
IV

Std dev, standard deviation

Efficacy

Objective response

- The primary study endpoint of OR by investigator assessment was achieved by 30 (50%) patients
- 29 (48.3%) patients achieved PR, one (1.7%) patient had a CR, and 20 (33.3%) had stable disease
- Median duration of response was 13.8 months (95% CI: 9.16, 18.88)

Kaplan–Meier curve of duration of OR

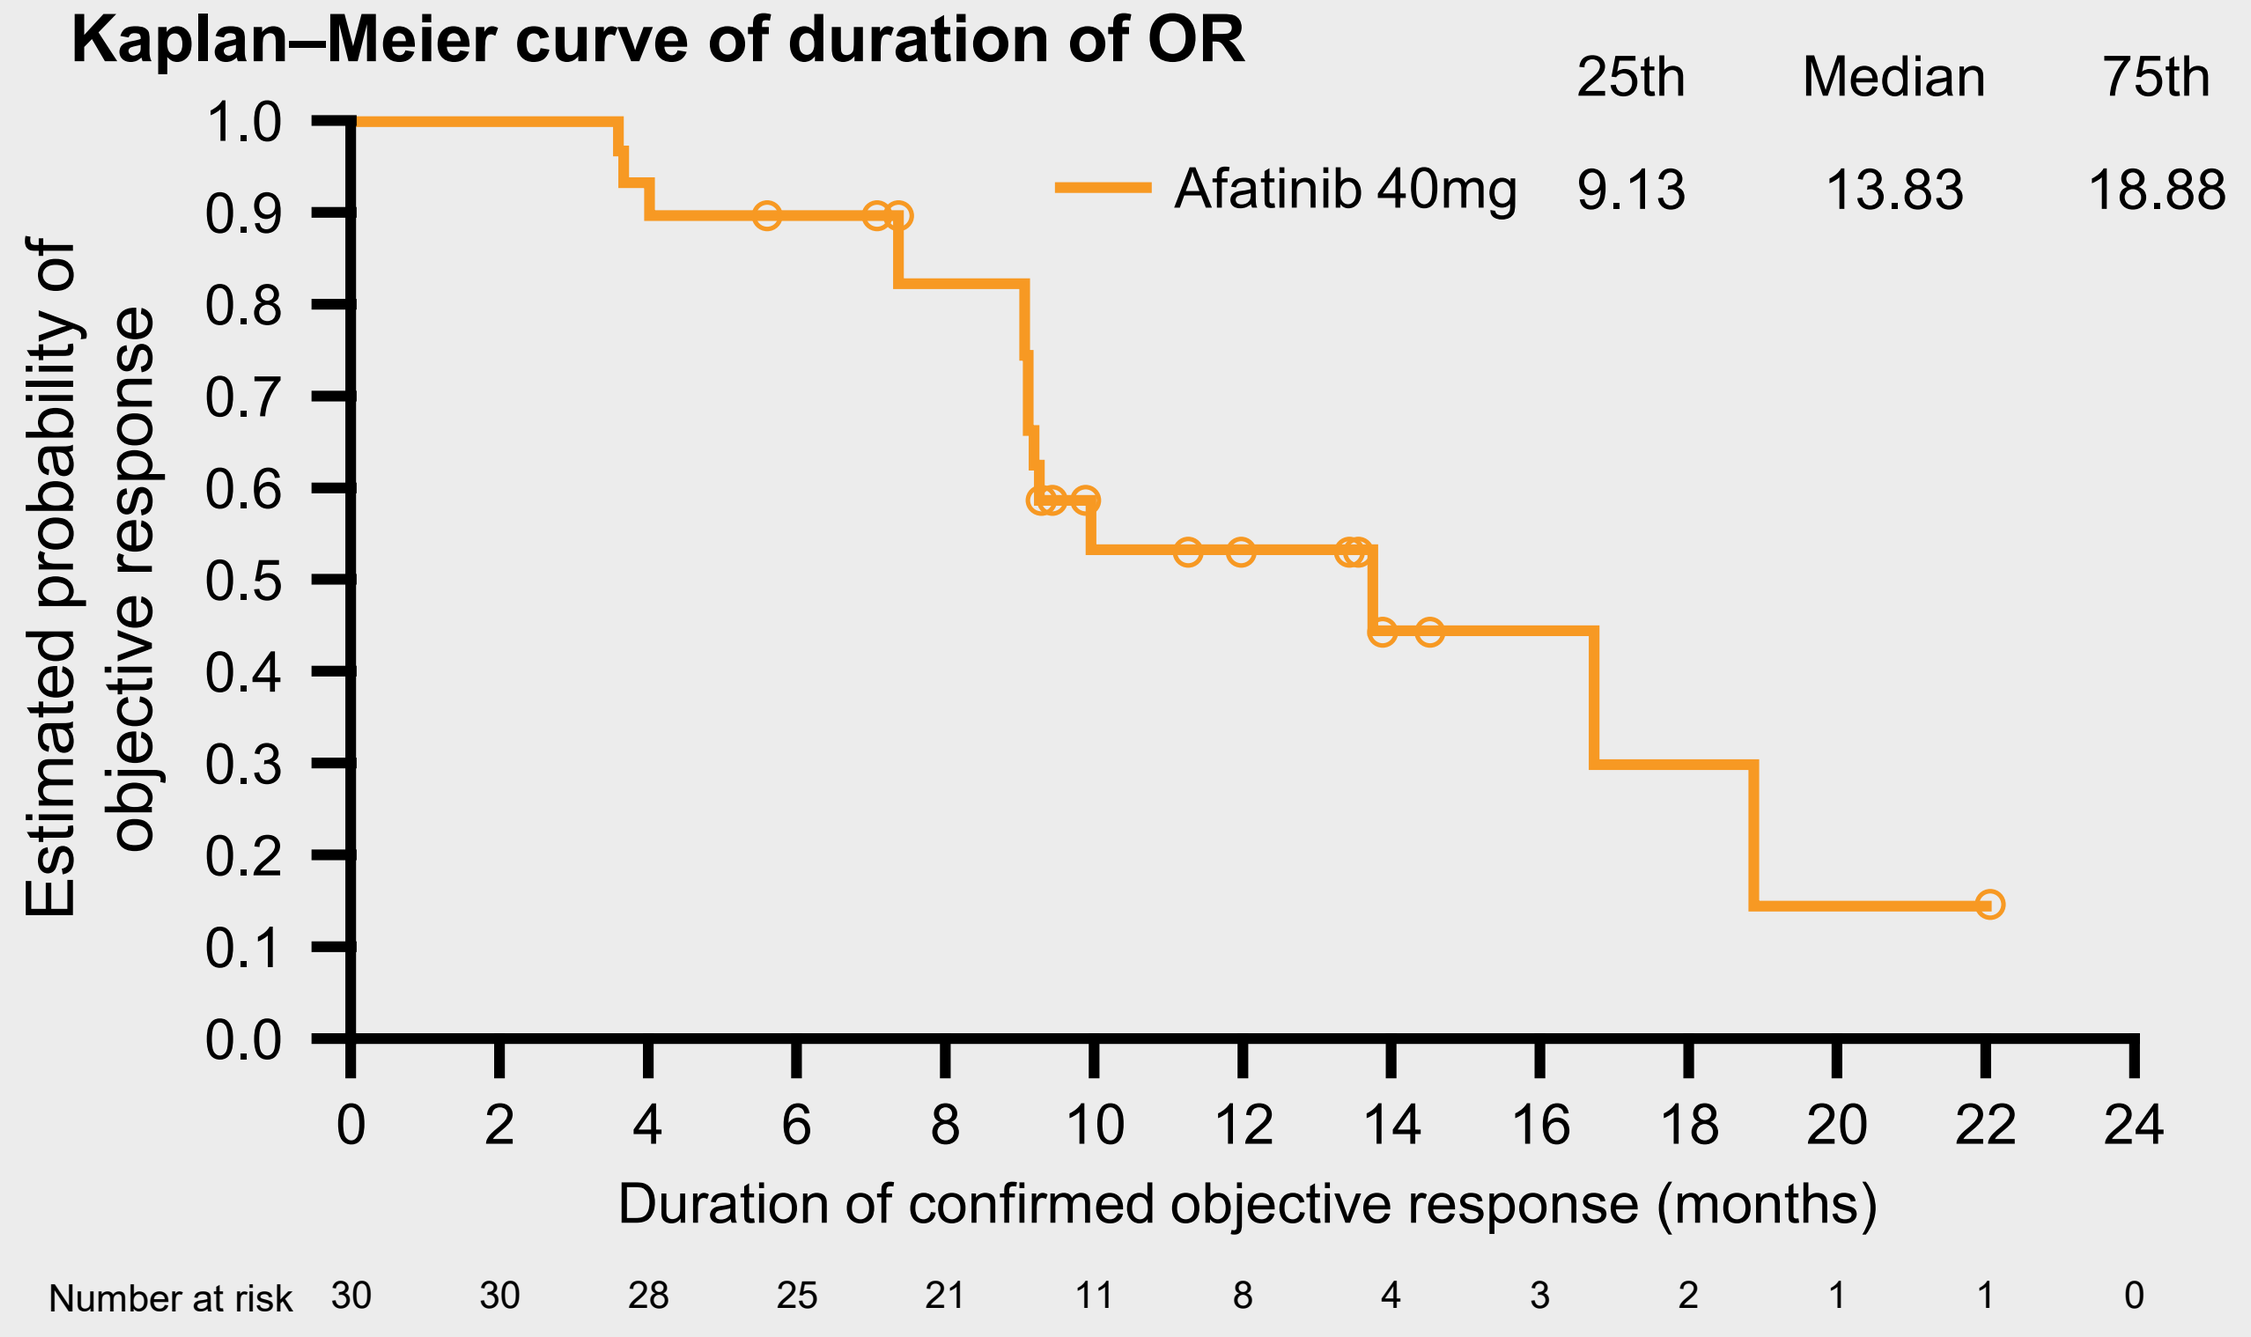
25th
Median
75th
Afatinib 40mg
9.13
13.83
18.88
Estimated probability of objective response
Duration of confirmed objective response (months)
Number at risk 30 30 28 25 21 11 8 4 3 2 1 1 0

CI, confidence interval

Efficacy (Cont'd)

Progression-free survival

- 39 patients (65.0%) experienced an event contributing to PFS analysis (i.e. disease progression as determined by investigator assessment, or death)
- Median PFS was 10.9 months (95% CI: 6.4, 13.2)

Kaplan–Meier curve of PFS

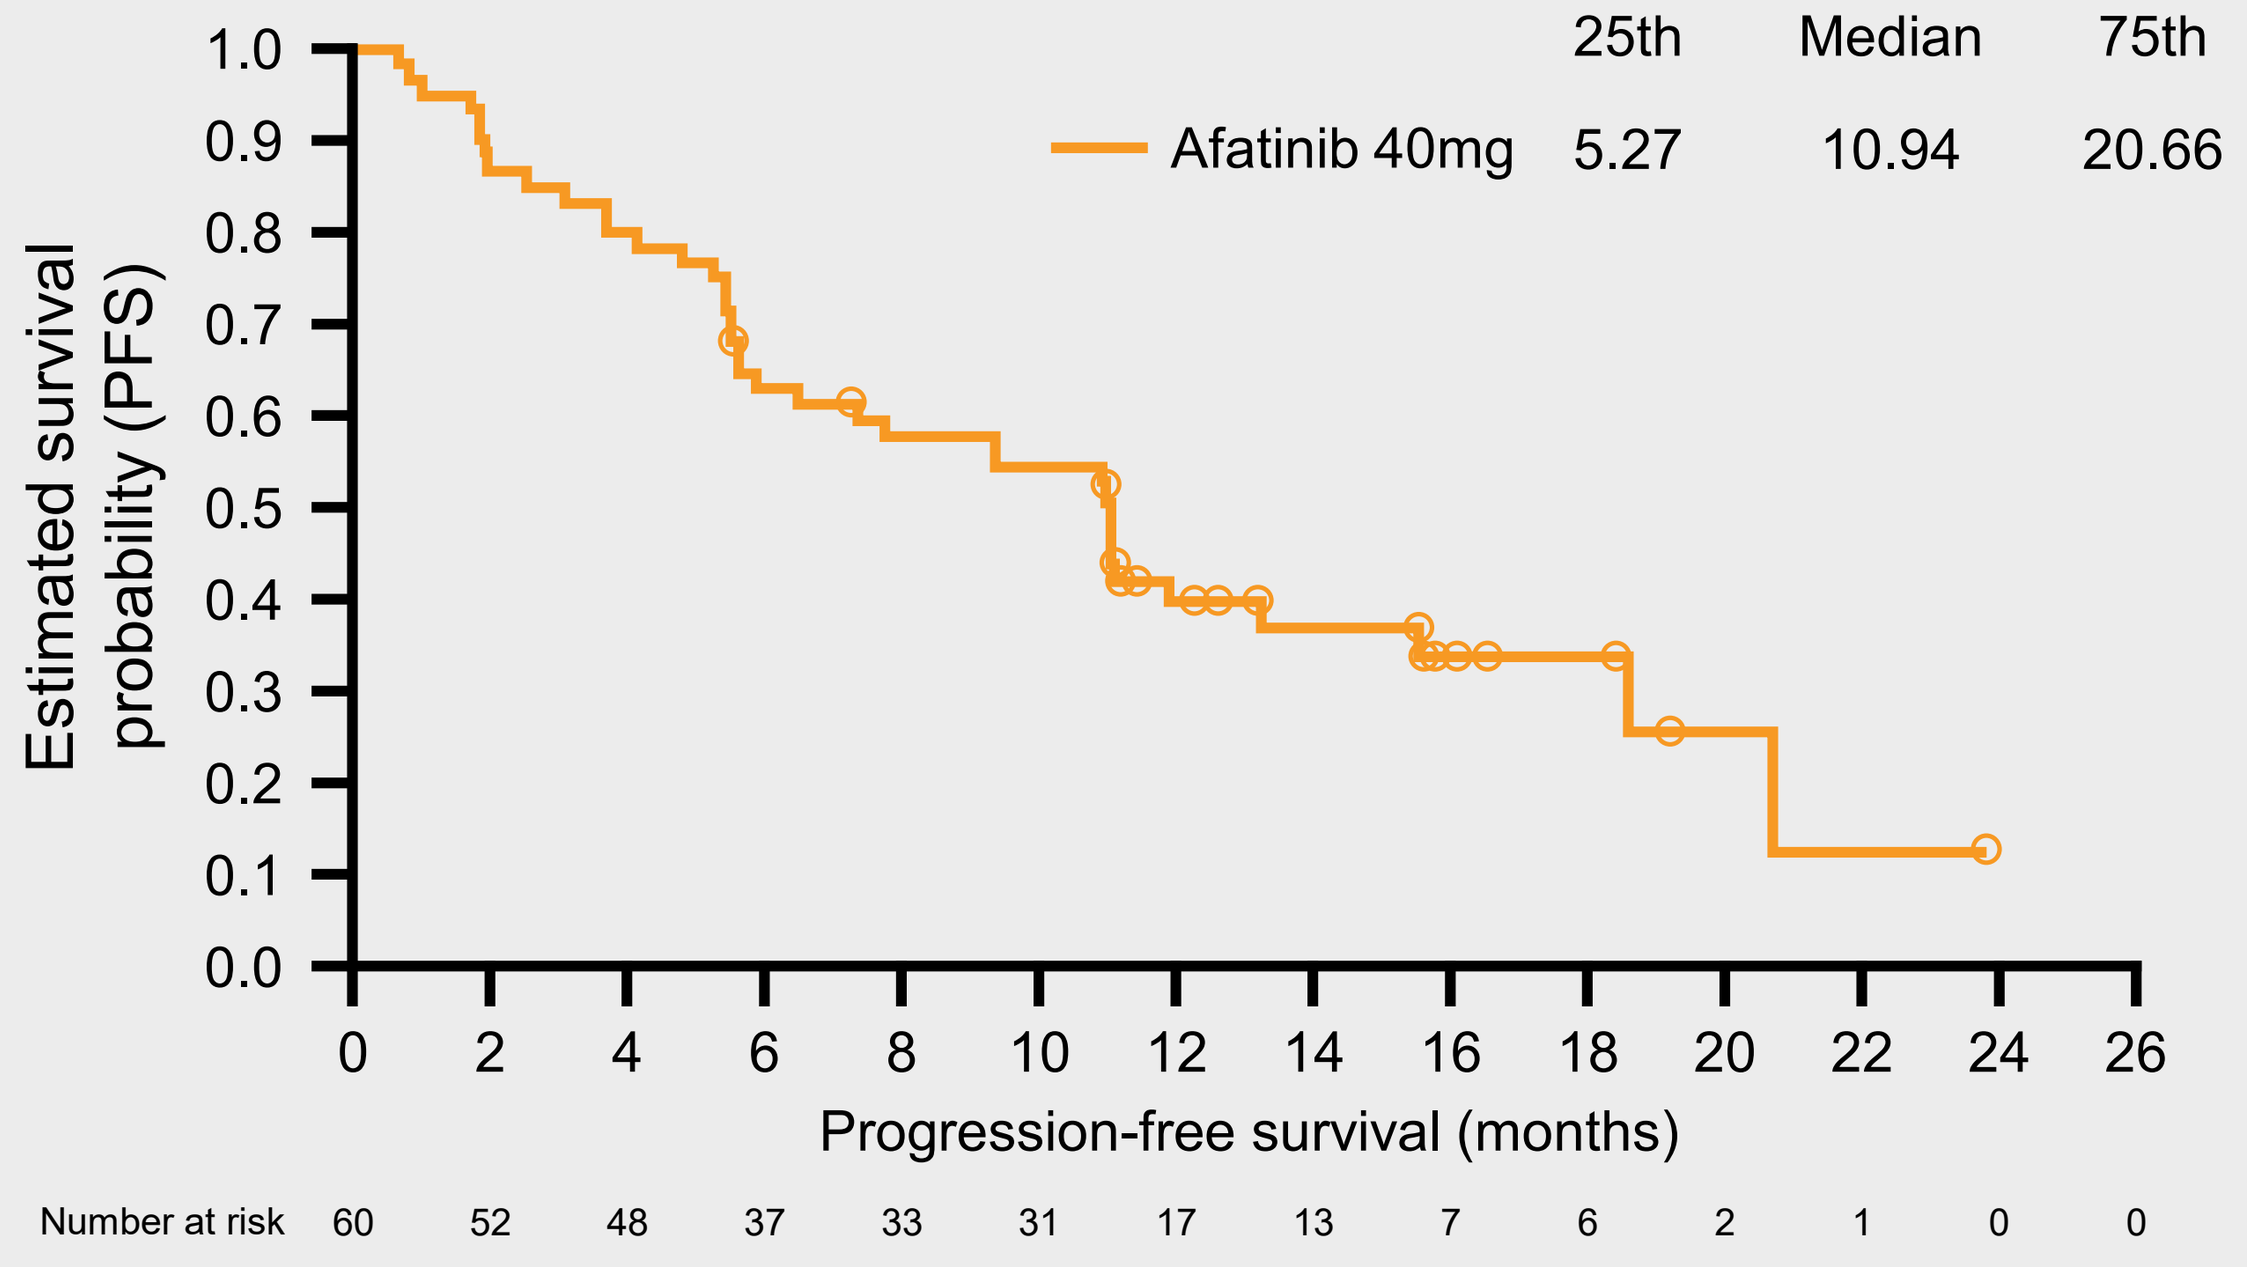
25th
Median
75th
Afatinib 40mg
5.27
10.94
20.66
Estimated survival probability (PFS)
Progression-free survival (months)
Number at risk 60 52 48 37 33 31 17 13 7 6 2 1 0 0

Disease control

- 50 patients (83.3%) showed confirmed disease control
- Median duration of disease control was 11.9 months (95% CI: 10.8, 20.7)

Kaplan–Meier curve of duration of disease control

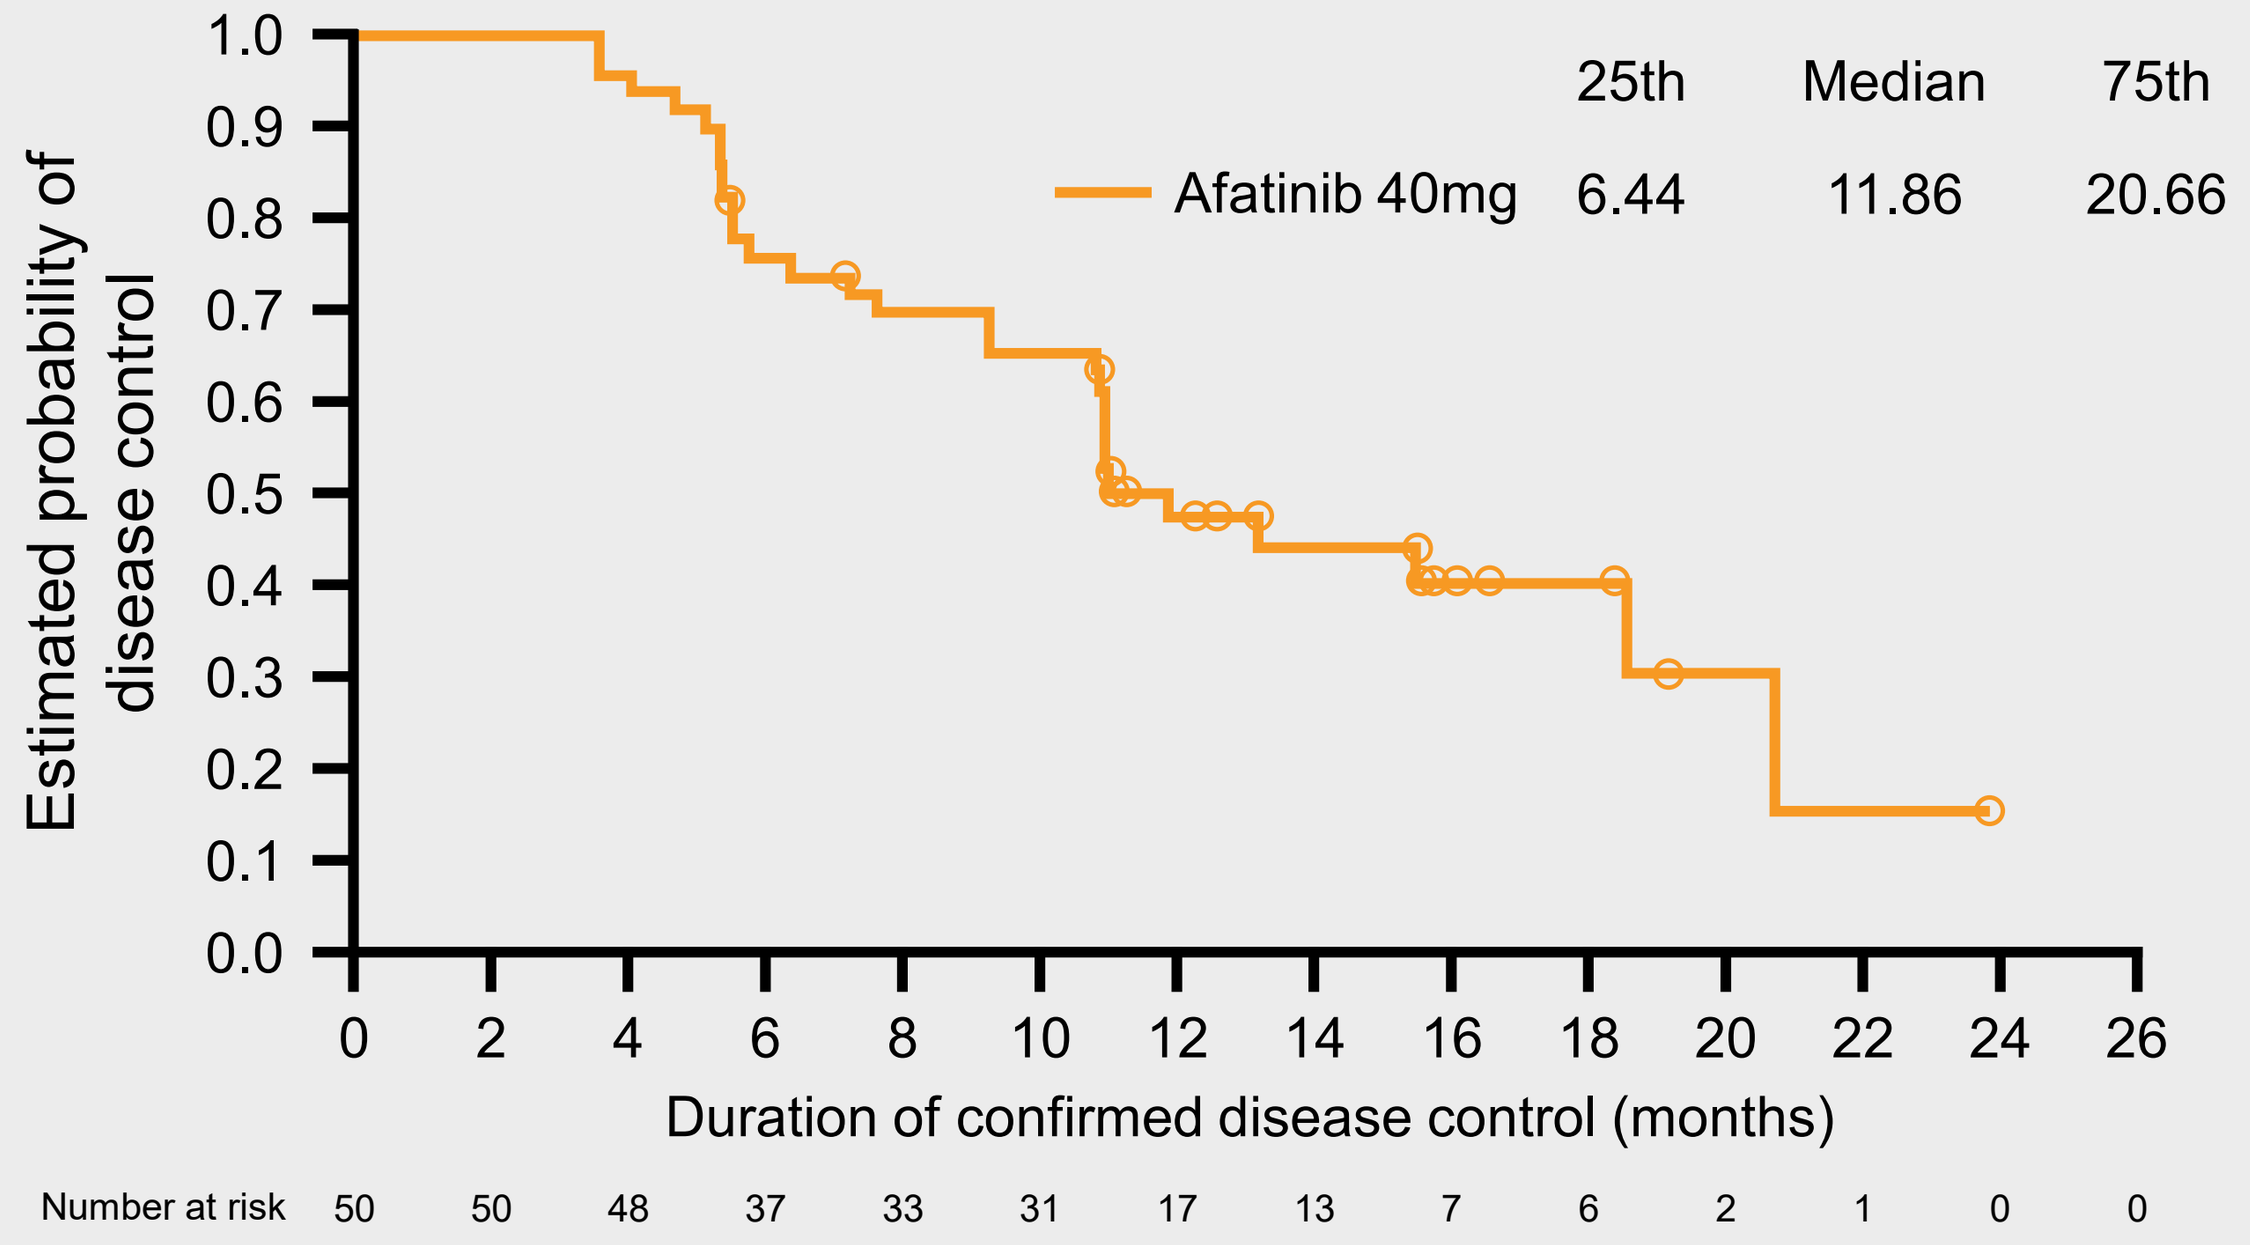
25th
Median
75th
Afatinib 40mg
6.44
11.86
20.66
Estimated probability of disease control
Duration of confirmed disease control (months)
Number at risk 50 50 48 37 33 31 17 13 7 6 2 1 0 0

Safety

- Most patients (91.7%) had at least one AE of any grade deemed to be treatment-related by the investigator
- The most commonly occurring drug-related AEs of any grade were diarrhoea (71.7%), rash (28.3%), paronychia (23.3%), mucosal inflammation (18.3%) and dermatitis acneiform (16.7%)
- 25 patients (41.7%) had dose reduction to 30 mg/day, with six patients (10.0%) having a further dose reduction to 20 mg/day
- 12 patients (20%) experienced AEs that led to permanent discontinuation of afatinib

Summary of AEs

	Afatinib 40 mg/day	
Patients	**n**	**%**
Any AE	57	95.0
Afatinib-related AEs	55	91.7
AEs leading to afatinib dose reduction	25	41.7
AEs leading to afatinib discontinuation*	12	20.0
Other significant AEs (according to ICH E3†)	25	41.7
Serious AEs‡	21	35.0
AEs by highest CTCAE grade		
Grade 1 or 2	32	53.3
Grade ≥3	25	41.7

*16 patients had AEs leading to discontinuation, but the reason for discontinuation in 4 patients was reported as PD according to RECIST or as clinical signs and symptoms of progression; †Guideline from the International Conference on Harmonisation of Technical Requirements for Registration of Pharmaceuticals for Human Use (ICH) on the structure and content of clinical study reports; ‡Patients could be counted in more than one category of serious AE.

Summary

- 50% of patients achieved a confirmed OR, with a median duration greater than 1 year; median PFS was 10.9 months
- More than 80% of patients had disease control, with a median duration of 11.9 months
- The safety and tolerability profile of afatinib was consistent with the known safety profile of afatinib, with the most common afatinib-related AEs observed being diarrhoea and rash
- The current study supports the use of afatinib as second-line therapy at the recommended 40 mg/day starting dose in EGFR TKI-naïve patients with locally advanced/metastatic NSCLC harboring common *EGFR* mutations (Del19 or L858R), after failure of first-line chemotherapy

References

1. Sequist LV, et al. J Clin Oncol 2013;31:3327–34
2. Wu Y-L, et al. Lancet Oncol 2014;15:213–22
3. Park K, et al. Lancet Oncol 2016;17:577–89
4. Yang JC-H, et al. Lancet Oncol 2012;13:539–48

Scan the QR code for an electronic copy of the poster and supplementary content†

†These materials are for personal use only and may not be reproduced without written permission of the authors and the appropriate copyright permissions.

Presented at the European Lung Cancer Congress (ELCC), Geneva, Switzerland, April 11–14, 2018

This study was funded by Boehringer Ingelheim. The authors were fully responsible for all content and editorial decisions, were involved at all stages of poster development and have approved the final version.
Medical writing assistance, supported financially by Boehringer Ingelheim, was provided by Hashem Dbouk of GeoMed, an Ashfield company, part of UDG Healthcare plc, during the development of this poster.
*Corresponding author email address: sumitra95@gmail.com