Efficacy and safety of nintedanib and docetaxel in lung adenocarcinoma patients following treatment with immune checkpoint inhibitors: first interim results of the ongoing non-interventional VARGADO study (NCT02392455)

55P

Abstract 389

Christian Grohé,[1] Wolfgang Gleiber,[2] Siegfried Haas,[3] Harald Müller-Huesmann,[4] Mathias Schulze,[5] Judith Atz,[6] Rolf Kaiser[6]

[1]Department of Pneumology, ELK, Berlin, Germany; [2]University Hospital Frankfurt, Pneumology/Allergology, Frankfurt, Germany; [3]Friedrich-Ebert Hospital Neumuenster, Clinics for Haematology, Oncology and Nephrology, Neumuenster, Germany; [4]Klinik für Hämatologie und Onkologie, Bruederkrankenhaus St. Josef, Paderborn, Germany; [5]Praxis Dr. Schulze, Zittau, Germany; [6]Medical Affairs Oncology, Boehringer Ingelheim Pharma GmbH & Co KG, Ingelheim Am Rhein, Germany

INTRODUCTION

- Nintedanib is an oral, triple angiokinase inhibitor targeting vascular endothelial growth factor (VEGF) receptors 1–3, platelet-derived growth factor (PDGF) receptors α/β, and fibroblast growth factor (FGF) receptors 1–3,[1] as well as RET[2]
- Nintedanib is approved in the EU and other countries in combination with docetaxel for the treatment of patients with locally advanced, metastatic or locally recurrent non-small cell lung cancer (NSCLC) of adenocarcinoma histology after first-line chemotherapy[3]
- Approval was based on data from the Phase III LUME-Lung 1 trial, in which nintedanib plus docetaxel was associated with a clinically meaningful extension in overall survival (OS) compared with placebo plus docetaxel in patients with adenocarcinoma NSCLC who had progressed after first-line chemotherapy[4]
- Following the introduction of second-line immune checkpoint inhibitors (ICIs), the treatment landscape in non-targetable advanced NSCLC is continuing to advance with their use in the first-line setting[5]
- With this growing number of treatment options, it is becoming increasingly important in clinical practice to establish the optimal treatment after ICI ± chemotherapy. So far, none of the available treatments have been investigated in patients who have progressed on ICIs
- An immunosuppressive tumour microenvironment (TME) is closely linked with hypoxia-induced, VEGF-promoted angiogenesis and upregulation of immunosuppressive cells.[6] It is possible that the abnormal vasculature resulting from tumour angiogenesis contributes to or underlies some tumours' resistance to ICIs. This same mechanism would also prime the tumour for anti-angiogenic therapy
- We hypothesise that an anti-angiogenic treatment strategy involving inhibition of VEGF, as well as PDGF and FGF, tips the balance towards an immunosupportive TME: an 'Angio-Immunogenic Switch' **(Figure 1)**

Figure 1. 'Angio-Immunogenic Switch'

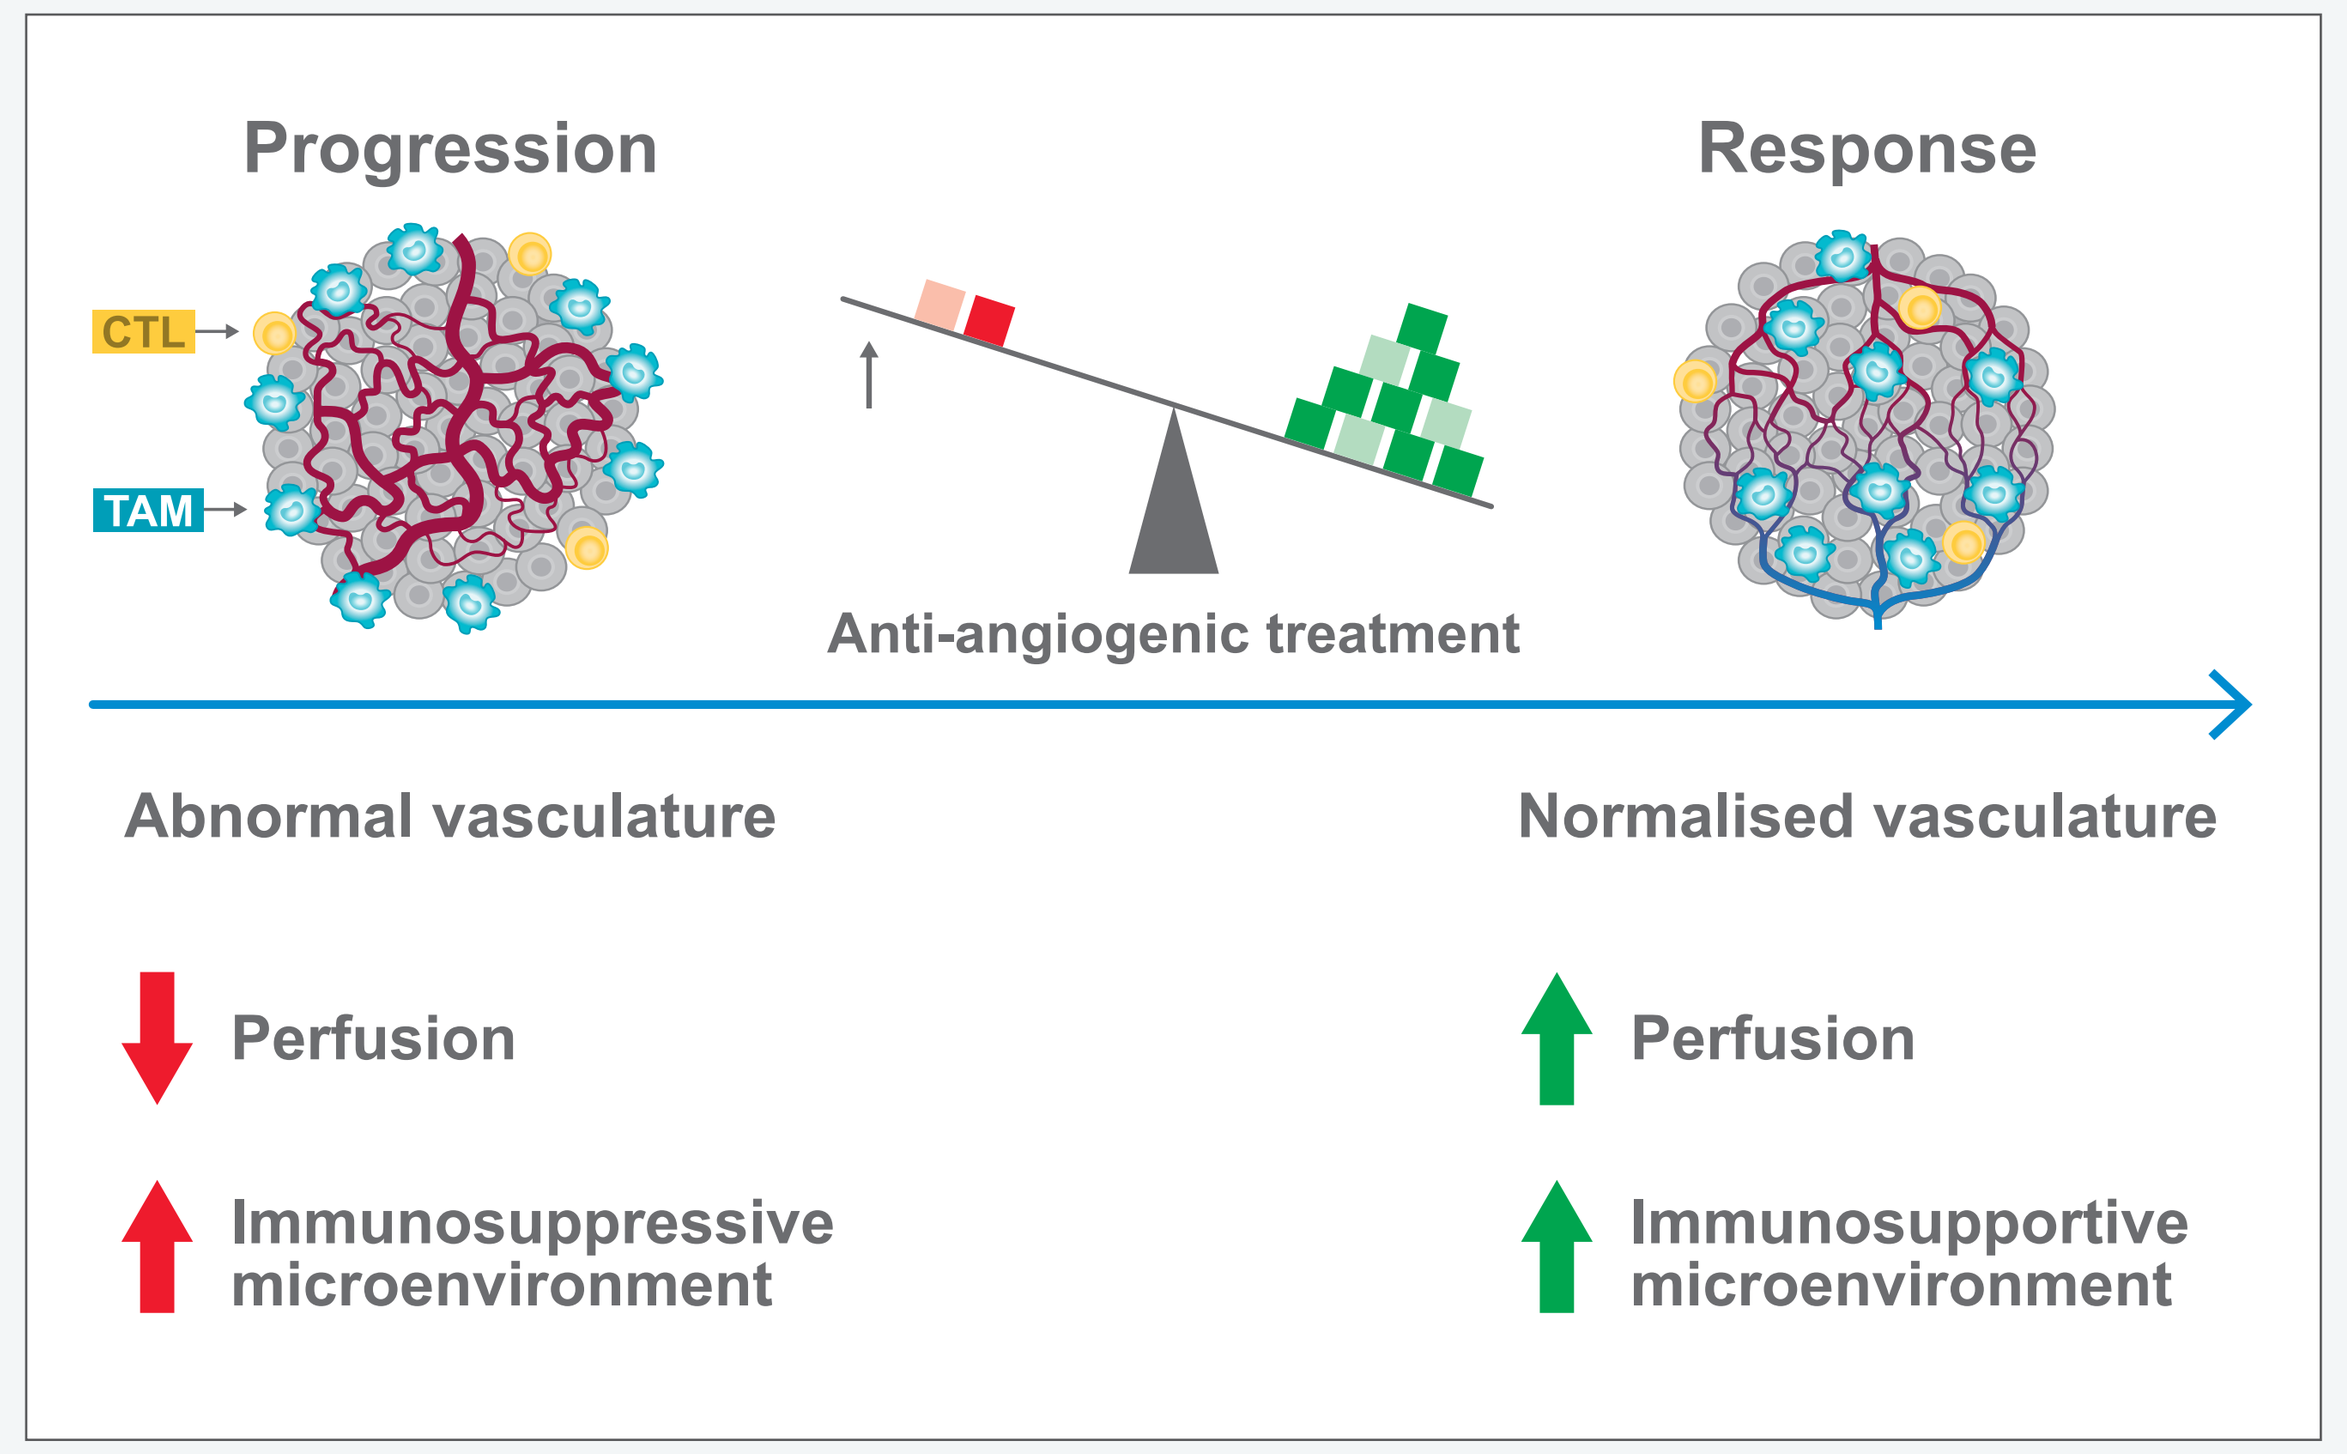
Progression
Response
CTL
TAM
Anti-angiogenic treatment
Abnormal vasculature
Normalised vasculature
Perfusion
Immunosuppressive microenvironment
Perfusion
Immunosupportive microenvironment

Modified from Fukumura D, et al. Nat Rev Clin Oncol 2018;15:325–40.
CTL, cytotoxic T lymphocyte; TAM, tumour-associated macrophage.

- One small dataset has been published on the efficacy of nintedanib plus docetaxel in patients with advanced lung adenocarcinoma who have previously been treated with ICIs: a retrospective analysis of patients in Spanish sites who participated in the nintedanib named patient use programme[7]
- In this dataset, 11 patients received nintedanib plus docetaxel after chemotherapy and ICI therapy. Treatment with nintedanib plus docetaxel was associated with an objective response rate (ORR) of 36%, a disease control rate (DCR) of 82% and a median progression-free survival (PFS) of 3.2 months[7]
- Here we present data from a larger cohort of patients who received nintedanib plus docetaxel after chemotherapy and ICI therapy in the non-interventional, prospective VARGADO study

STUDY DESIGN AND PATIENT POPULATION

- VARGADO is a prospective, non-interventional study of nintedanib plus docetaxel after first-line chemotherapy in the routine clinical treatment of patients with locally advanced, metastatic or locally recurrent adenocarcinoma NSCLC[8]
- Three patient cohorts in VARGADO are being evaluated (**Figure 2**)
- Between 15 March 2015 and 1 November 2018, 311 patients have been enrolled at 100 study sites across Germany
- We present the first interim analysis of Cohort B (N=22)

Figure 2. Patient cohorts in VARGADO

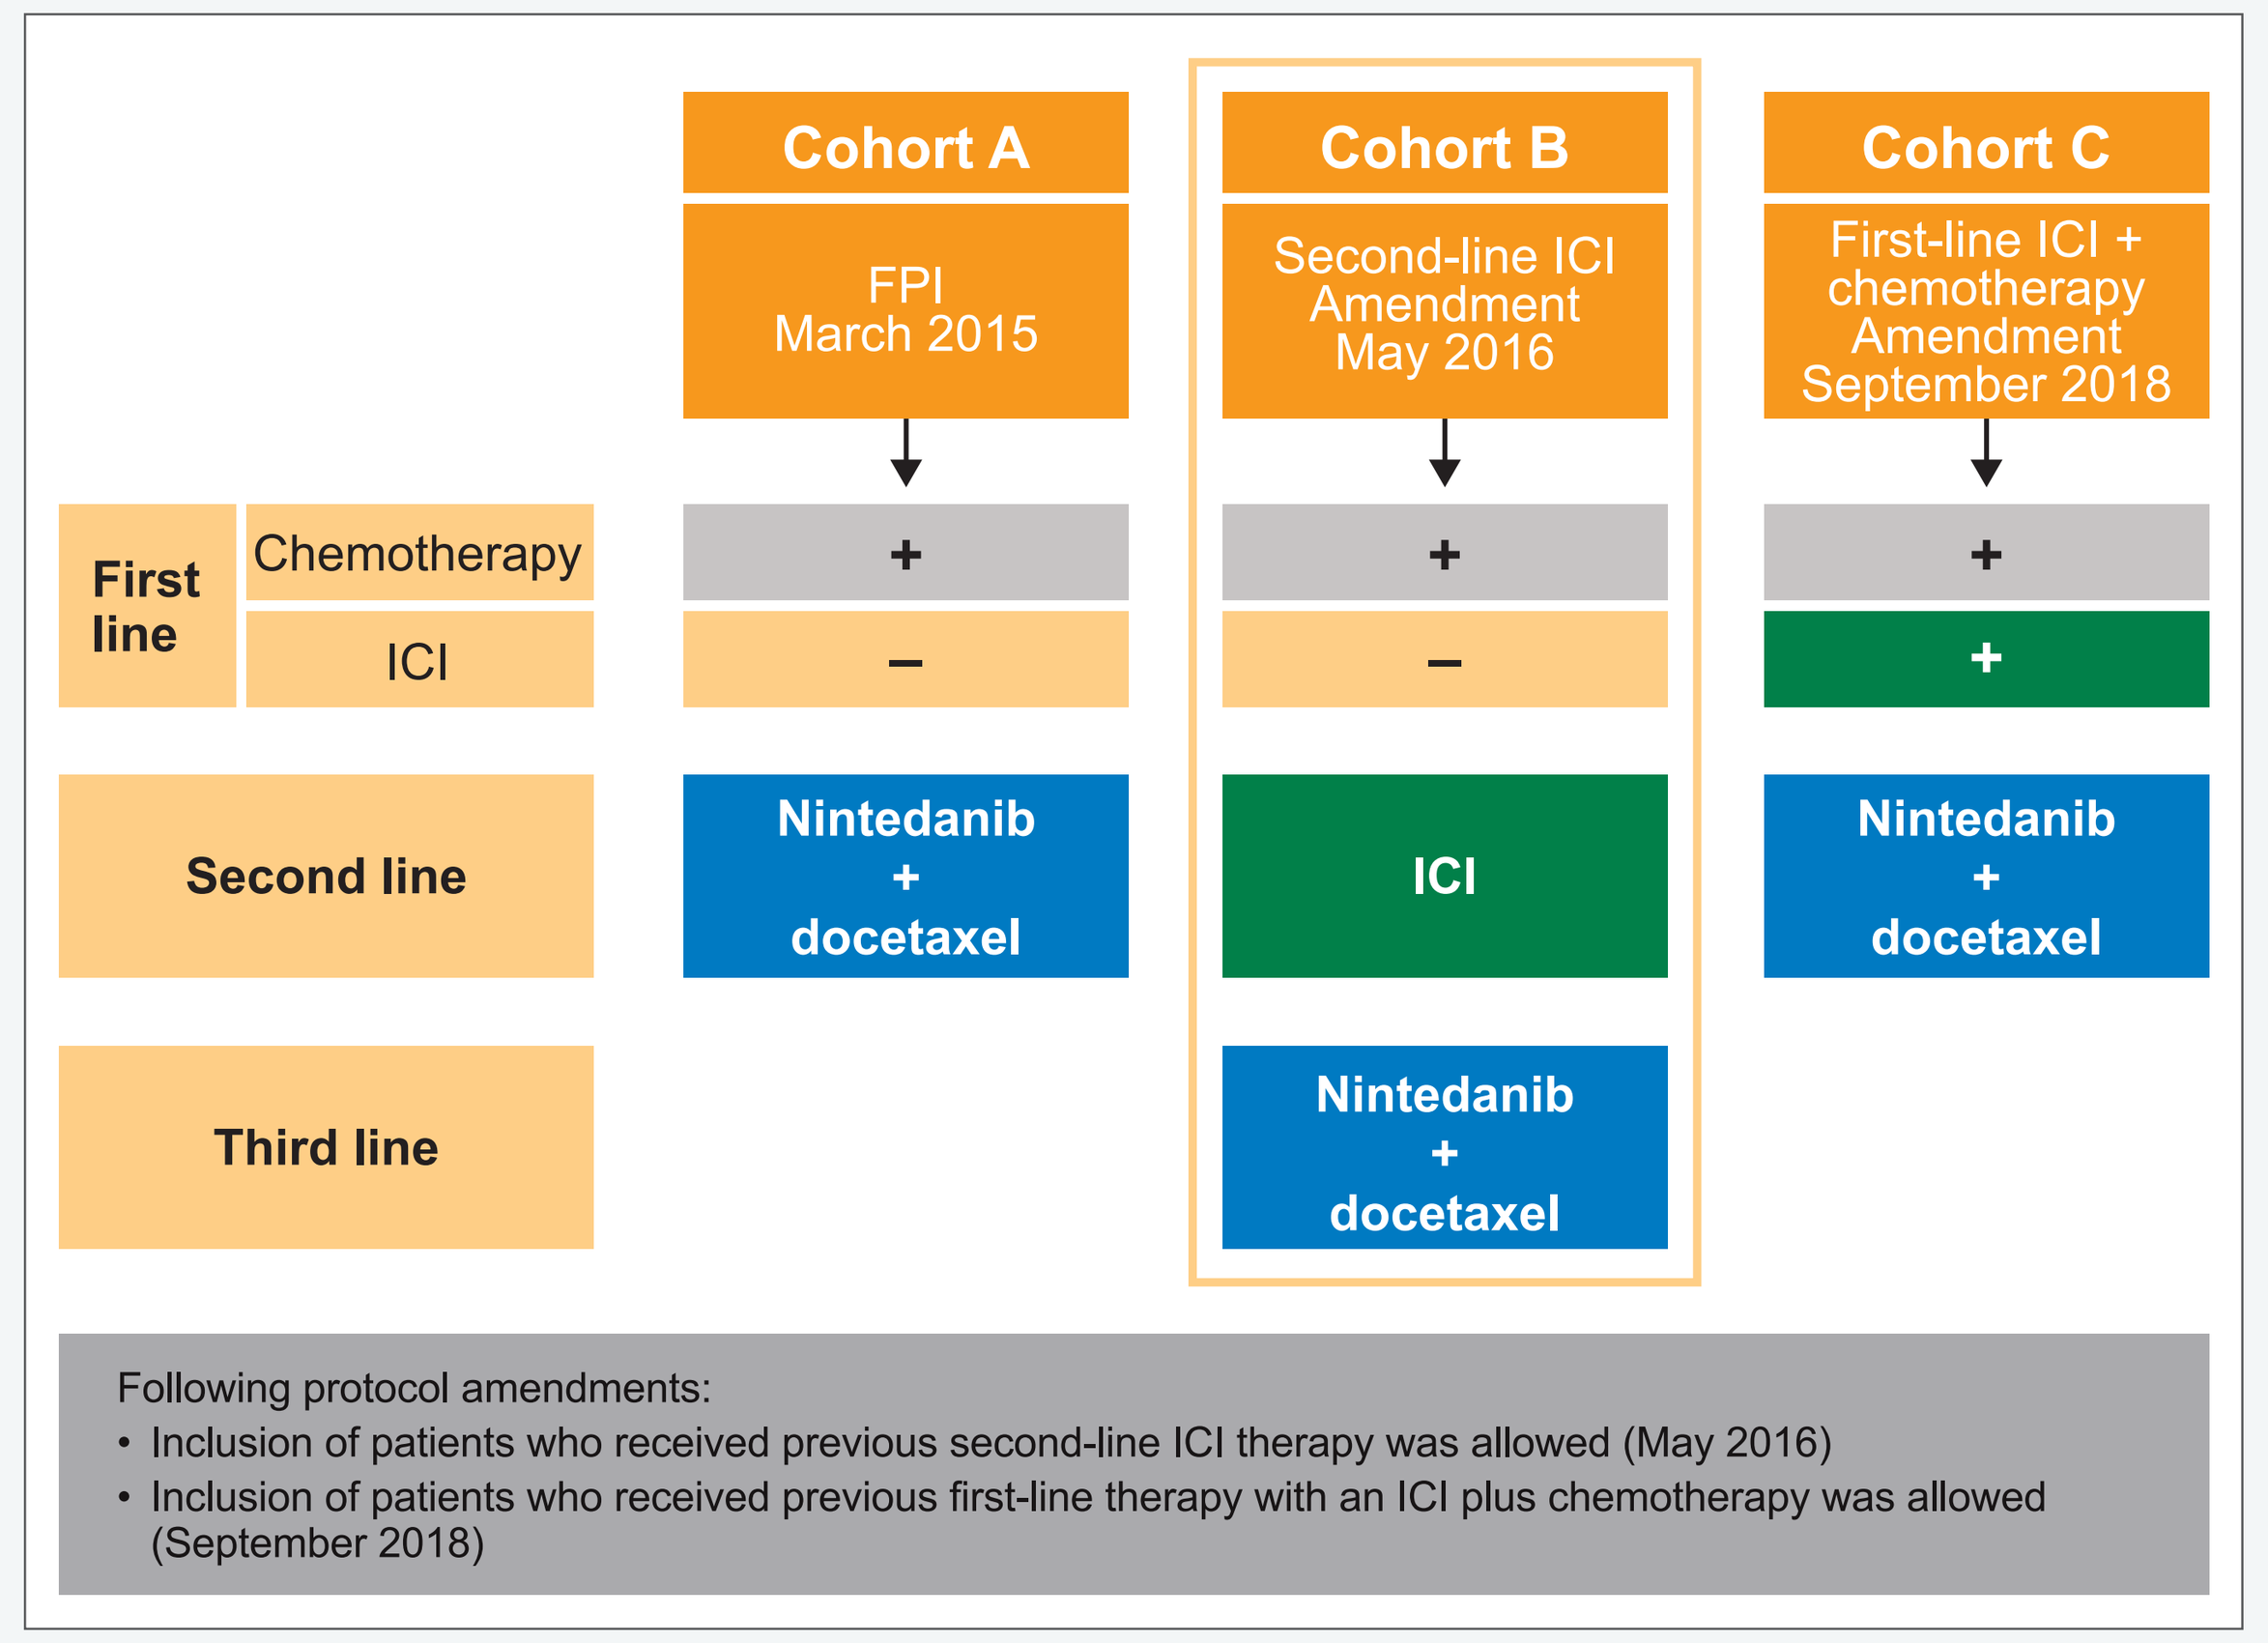
Cohort A
FPI
March 2015
Cohort B
Second-line ICI
Amendment
May 2016
Cohort C
First-line ICI + chemotherapy
Amendment
September 2018
First line
Chemotherapy
ICI
+
–
+
–
+
+
Second line
Nintedanib + docetaxel
ICI
Nintedanib + docetaxel
Third line
Nintedanib + docetaxel
Following protocol amendments:
• Inclusion of patients who received previous second-line ICI therapy was allowed (May 2016)
• Inclusion of patients who received previous first-line therapy with an ICI plus chemotherapy was allowed (September 2018)

FPI, first patient in; ICI, immune checkpoint inhibitor.

- Nintedanib and docetaxel were administered according to the approved label
- Patients were followed up for safety and efficacy for up to 24 months after the start of treatment. Patient data were collected during routine clinic visits
- All patients provided signed informed consent
- The primary endpoint is OS rate 1 year after the start of treatment with nintedanib plus docetaxel. Secondary endpoints include PFS, OS, ORR and DCR
- Incidence and severity of adverse events (AEs) are being reported according to the National Cancer Institute Common Terminology Criteria for Adverse Events, version 4.03

RESULTS

Patient characteristics

- Baseline characteristics for the 22 patients included in this analysis are shown in **Table 1**

Table 1. Baseline characteristics (N=22)

Median age, years (range)		58 (45–76)
Sex, % (n)	Male	68 (15)
ECOG PS, % (n)	0	18 (4)
	1	55 (12)
	2	9 (2)
	3	5 (1)
	Missing	14 (3)
Tumour stage at baseline, % (n)	IV	55 (12)
	<IV	18 (4)
	Unknown	14 (3)
	Missing	14 (3)
Brain metastases, % (n)	Yes	18 (4)
Previous first-line therapy,* % (n)	Pemetrexed	68 (15)
	Carboplatin	55 (12)
	Cisplatin	55 (12)
	Bevacizumab	27 (6)
	Vinorelbine	18 (4)
	Paclitaxel	9 (2)
	Docetaxel	5 (1)
Previous second-line therapy, % (n)	Nivolumab	77 (17)
	Pembrolizumab	23 (5)
Best response to second-line therapy, % (n)	ORR	23 (3/13§)
	SD	15 (2/13)
	PD	62 (8/13)
	Not documented, n	9

*Multiple concurrent first-line therapies possible; §Of which one was a CR.
CR, complete response; ECOG PS, Eastern Cooperative Oncology Group performance status; ORR, objective response rate; PD, progressive disease; SD, stable disease.

Efficacy

- At the time of this interim analysis (data cut-off: 1 August 2018), median duration of follow-up was 6.0 months (95% confidence interval [CI]: 3.5–8.8)
- 14 PFS events had occurred (10 patients had disease progression and four patients died). Seven patients were censored and data were not yet available for one patient
- Median PFS was 5.5 months (95% CI: 1.9–8.7; **Figure 3**)

Figure 3. PFS by investigator assessment (n=21)

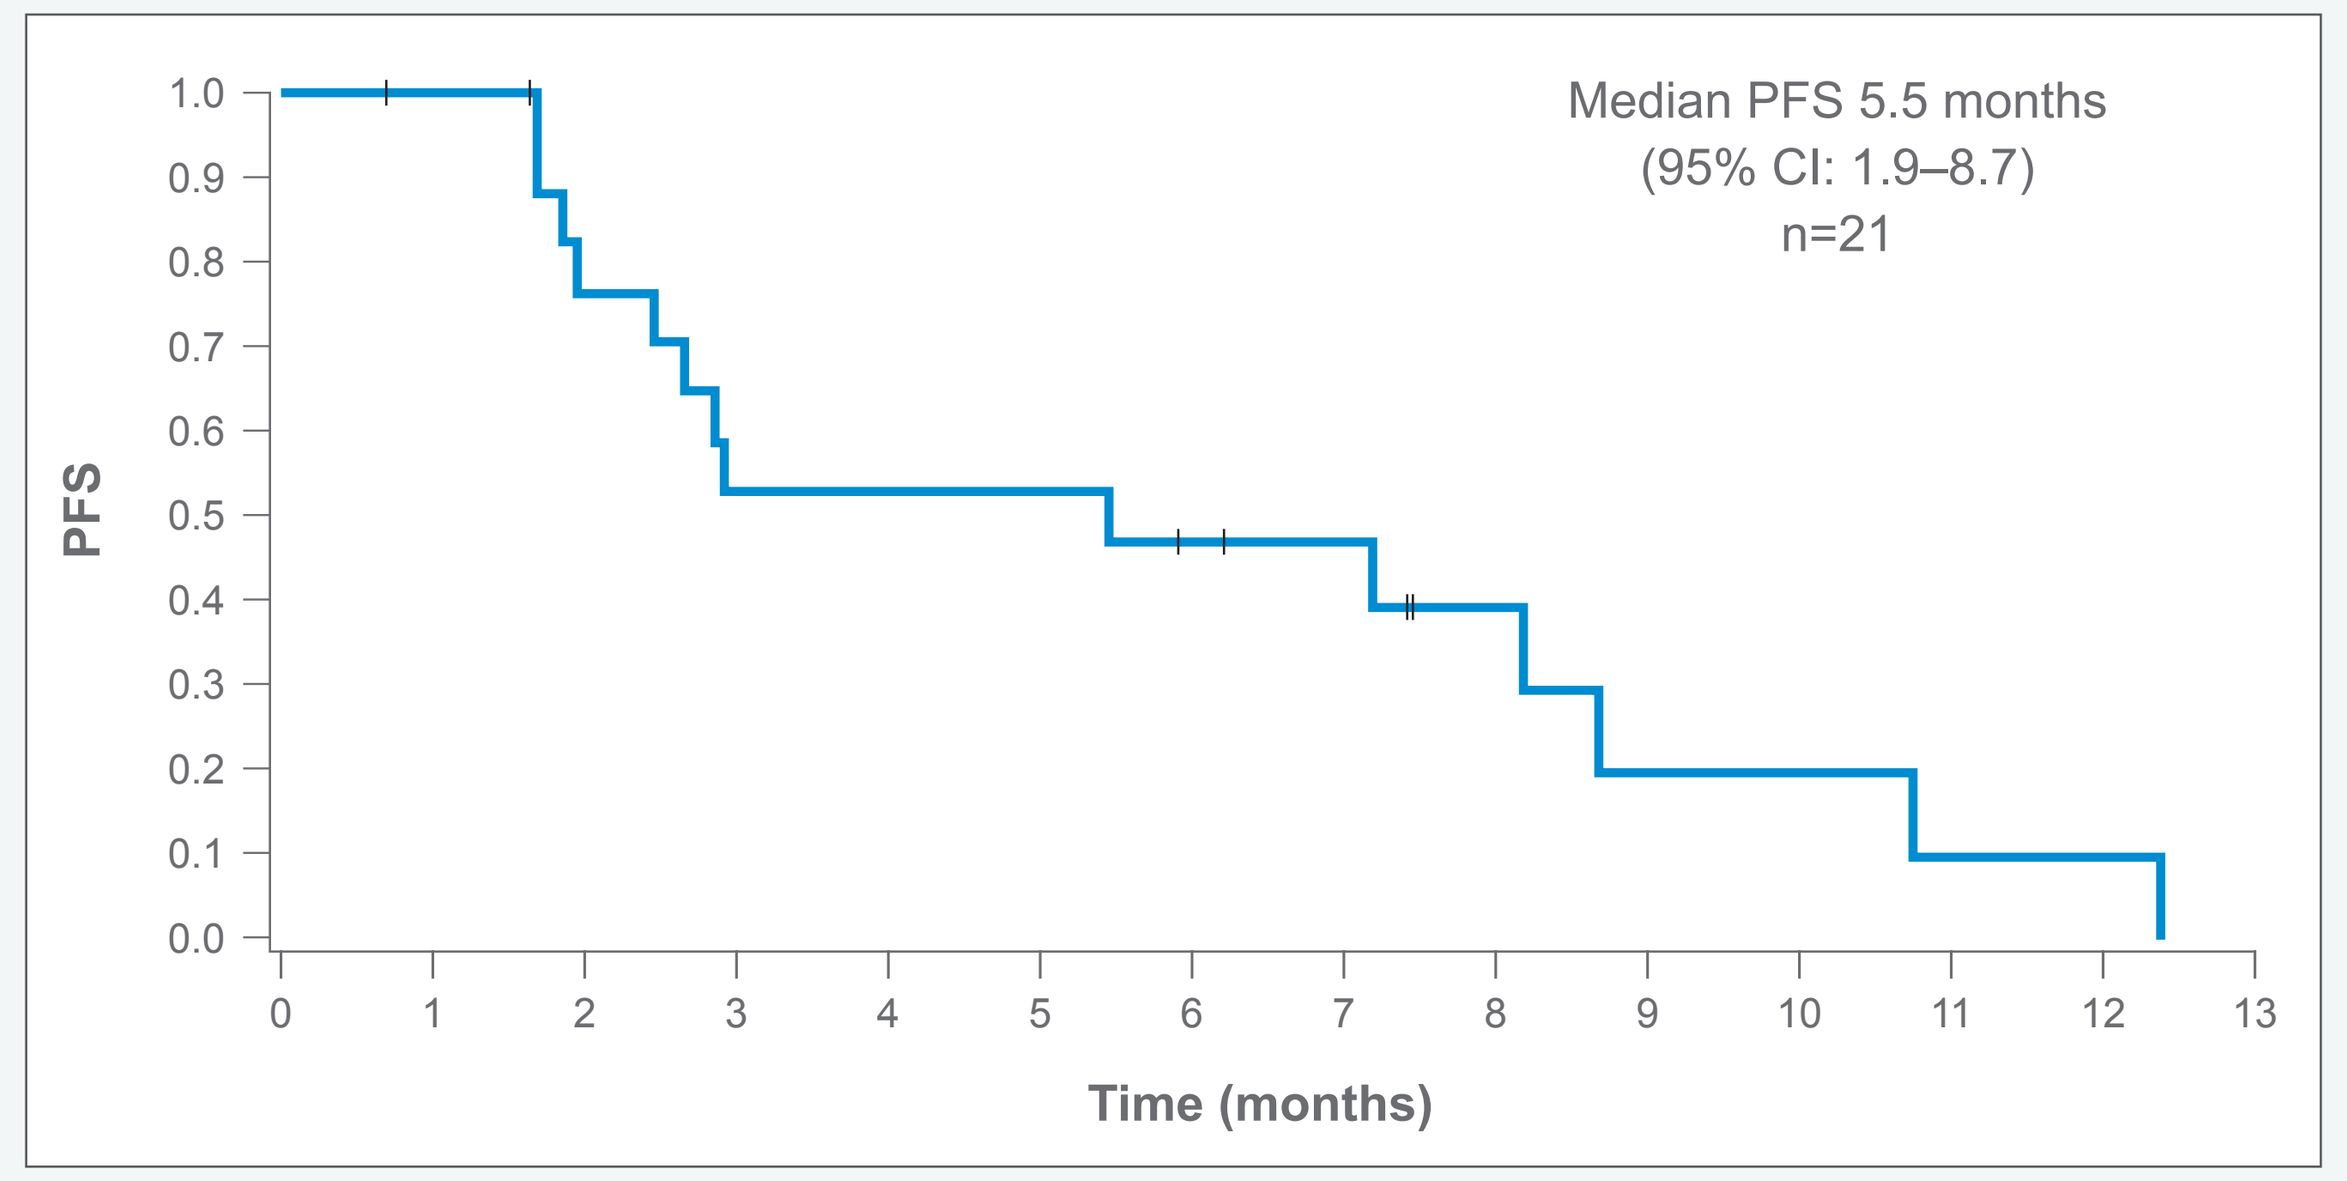
Median PFS 5.5 months
(95% CI: 1.9–8.7)
n=21
PFS
1.0
0.9
0.8
0.7
0.6
0.5
0.4
0.3
0.2
0.1
0.0
0 1 2 3 4 5 6 7 8 9 10 11 12 13
Time (months)

Data cut-off: 1 August 2018.
CI, confidence interval; PFS, progression-free survival.

- Treatment duration by treatment for each patient is shown in **Figure 4**

Figure 4. Swimmer plot showing treatment duration by treatment for each patient

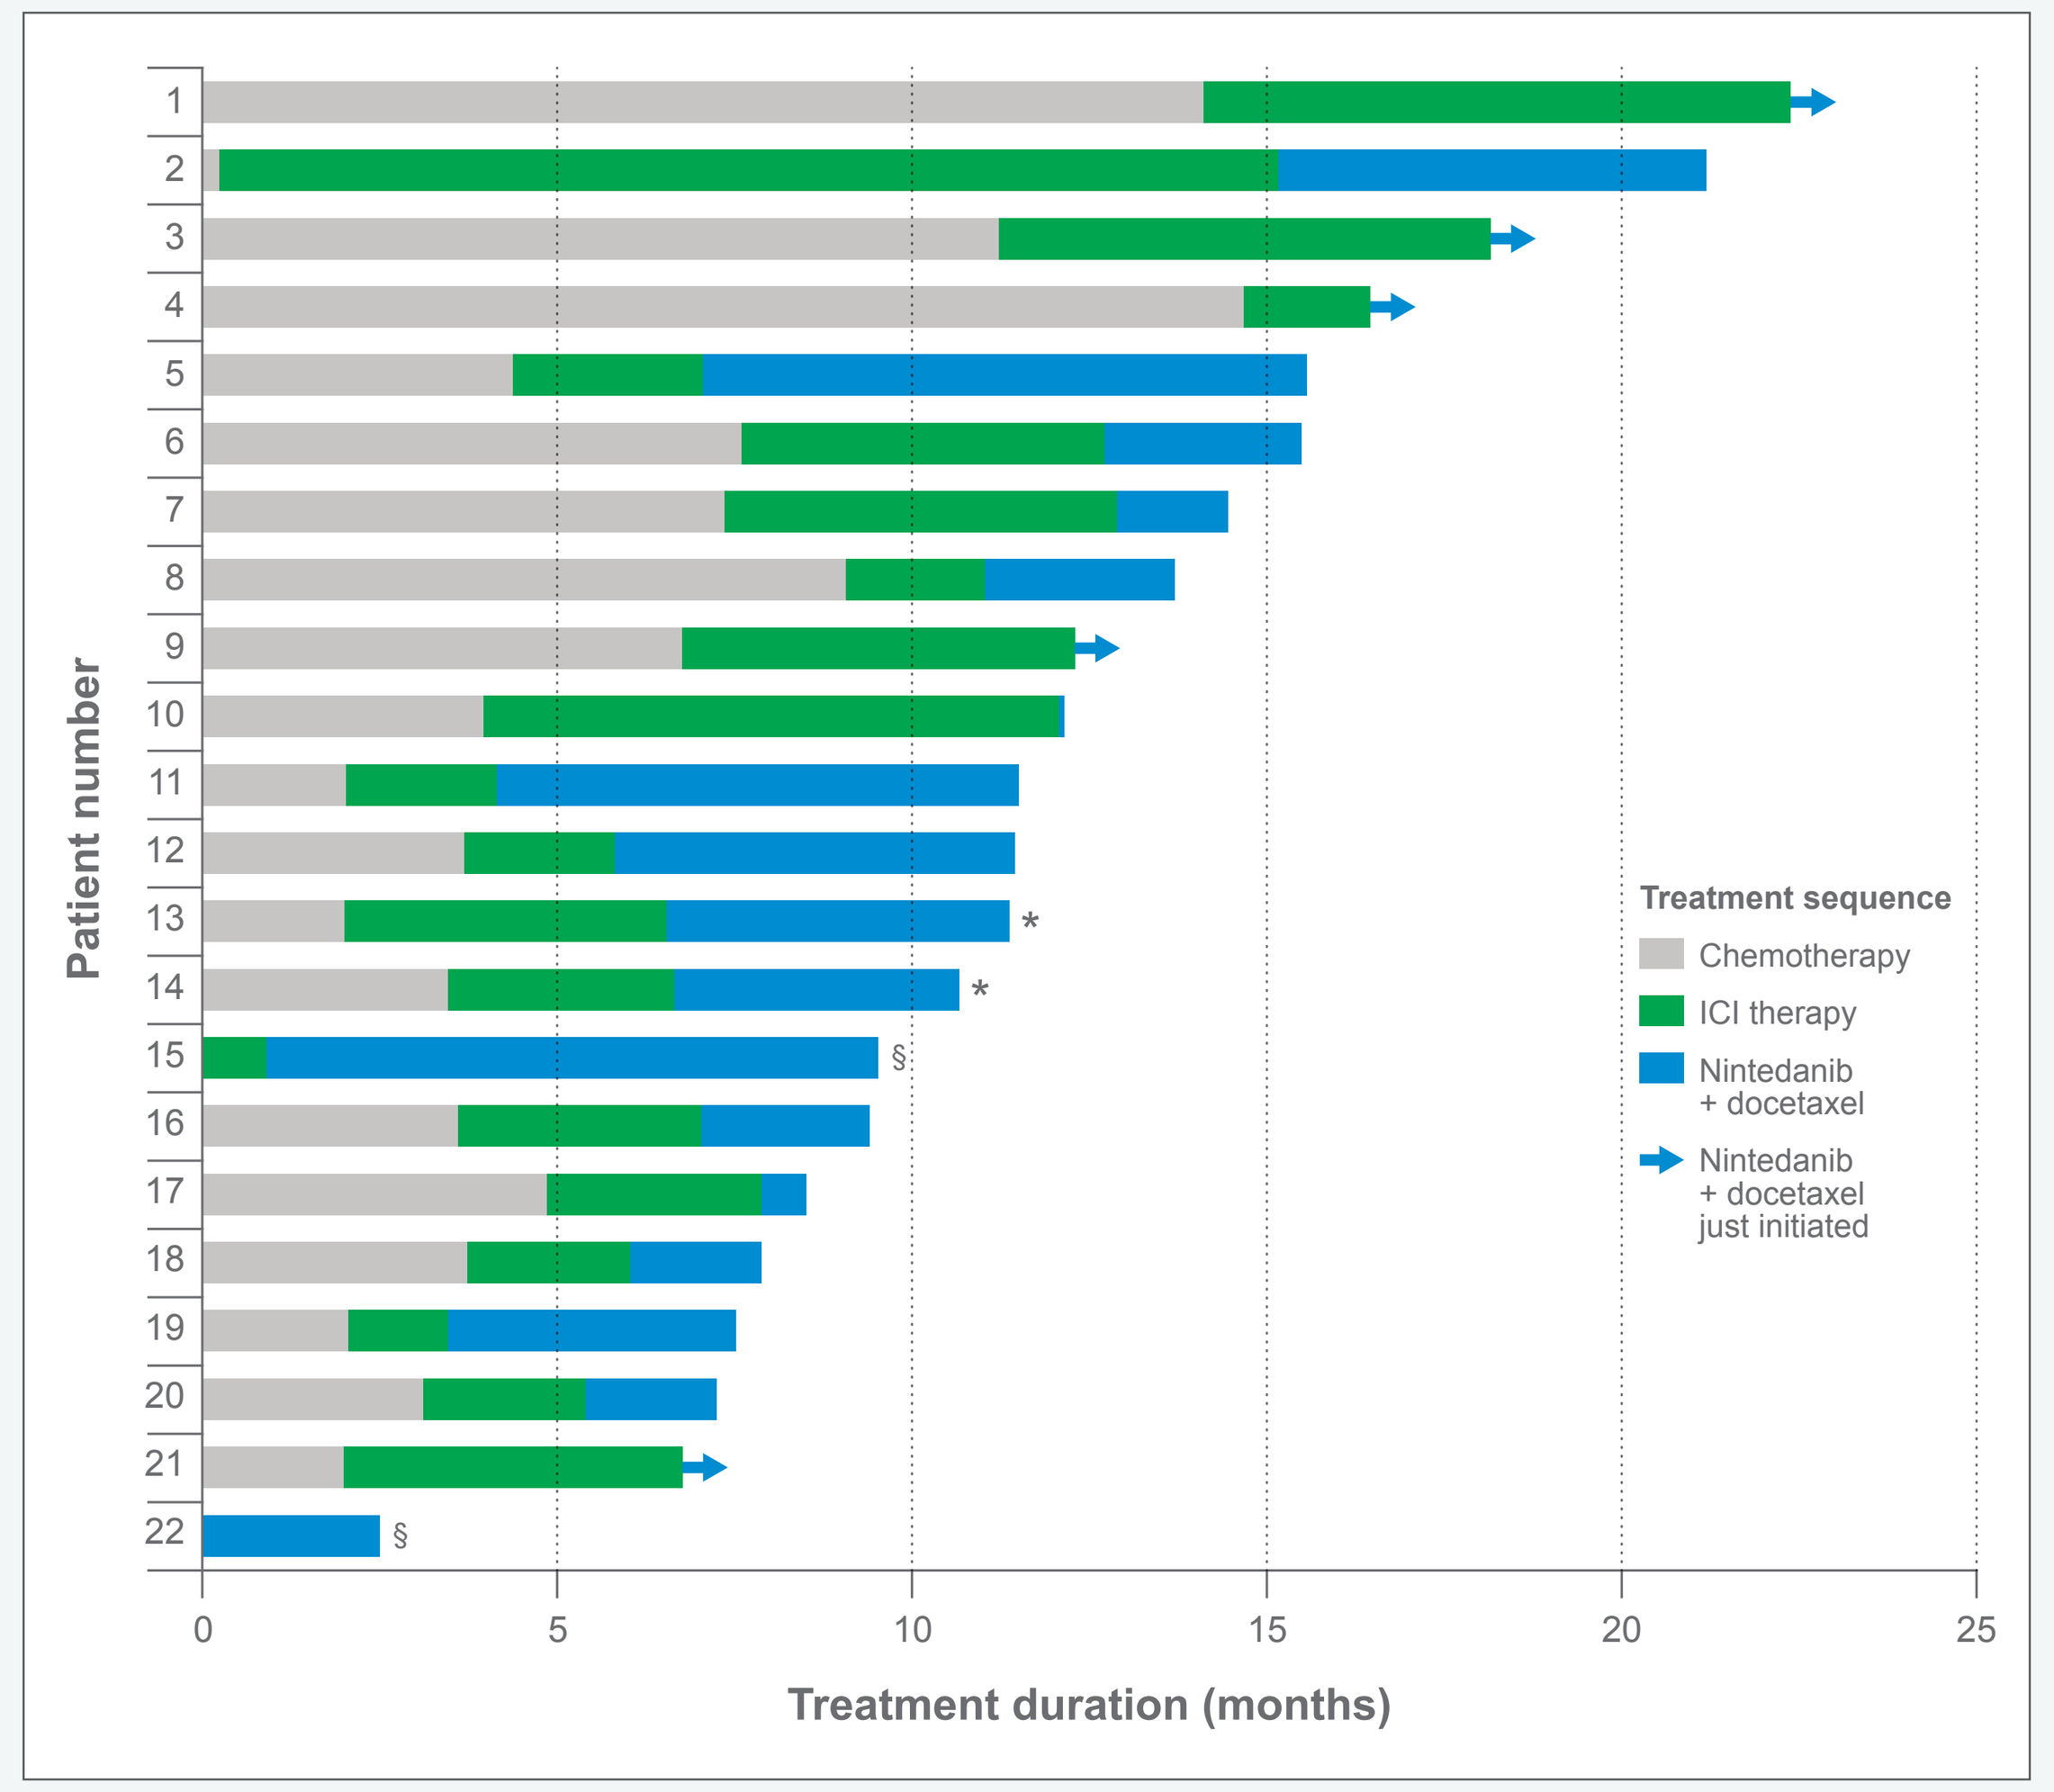
Patient number
1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16 17 18 19 20 21 22
0 5 10 15 20 25
Treatment duration (months)
Treatment sequence
Chemotherapy
ICI therapy
Nintedanib + docetaxel
Nintedanib + docetaxel just initiated

*Single-agent nintedanib treatment ongoing; §Previous therapies not documented.
Data cut-off: 1 August 2018. ICI, immune checkpoint inhibitor.

- OS data are not yet mature and are not reported here
- ORR data and DCR data (**Table 2**) were available for 12 patients at the time of analysis
- Seven patients (32%) had at least one nintedanib dose reduction and five patients (23%) had at least one docetaxel dose reduction

Table 2. Best response to nintedanib plus docetaxel (n=12)

ORR, % (n)	58 (7)
PR, % (n)	58 (7)
SD, % (n)	25 (3)
DCR, % (n)	83 (10)

DCR, disease control rate; ORR, objective response rate; PR, partial response; SD, stable disease.

Safety

- Safety was evaluated in all 22 patients
- 21 (95%) patients had AEs; 11 (50%) had serious AEs
- The most common nintedanib-related AEs of any grade were diarrhoea (36%), stomatitis (27%), decreased white blood cell count (14%) and nausea (14%)
- The most common Grade ≥3 nintedanib-related AEs were stomatitis (14%), decreased white blood cell count (14%) and febrile neutropenia (9%)
- The most common docetaxel-related AEs of any grade were diarrhoea (27%), stomatitis (27%), decreased white blood cell count (23%) and nausea (18%)
- The most common Grade ≥3 docetaxel-related AEs were decreased white blood cell count (18%), stomatitis (14%), and nausea, decreased neutrophil count and febrile neutropenia (each 9%)

CONCLUSIONS

- These data, while from a small patient cohort, provide a second independent indication that patients who progress on previous ICI therapy may benefit from treatment with nintedanib plus docetaxel
- We propose that the rational sequencing of anti-angiogenic therapy after ICIs may be a promising approach to treatment in this patient population that warrants further investigation
- Further research into a potential 'Angio-Immunogenic Switch' may facilitate patient selection and optimal treatment sequencing in future
- Collection of additional clinical data in this area is ongoing, with patient recruitment in the VARGADO study being extended

REFERENCES

1. Hilberg F, et al. Cancer Res 2008;68:4774–82.
2. Boehringer Ingelheim. Data on file.
3. Vargatef® Summary of Product Characteristics. http://www.ema.europa.eu/ docs/en_GB/document_library/EPAR_-_Product_Information/human/002569/WC500179970.pdf (Accessed: 6 September 2018).
4. Reck M, et al. Lancet Oncol 2014;15:143–55.
5. Planchard D, et al. Ann Oncol 2018;29(Suppl. 4):iv192–237.
6. Fukumura D, et al. Nat Rev Clin Oncol 2018;15:325–40.
7. Corral, J et al. Poster presented at WCLC 2017 (Poster P2.01-022).
8. Grohé C, et al. Poster presented at ESMO 2018 (Poster 1504TiP).

ACKNOWLEDGEMENTS

Disclosures: The authors were fully responsible for all content and editorial decisions, were involved in all stages of poster development, and have approved the final version.

During the preparation of this poster, medical writing assistance, supported financially by Boehringer Ingelheim, was provided by Syneos Health, UK.

Disclaimer: Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without written permission from the authors.

To download a copy of the poster, scan the QR code or type the following URL into your browser: https://bit.ly/2DRRGOo

Corresponding author: Rolf Kaiser (rolf.kaiser@boehringer-ingelheim.com)

Presented at the European Society for Medical Oncology Immuno-Oncology (ESMO I-O) Congress, Geneva, Switzerland, 13–16 December 2018